415

HAND Improvement is Associated with Increase CPE score After ARV Intensification - *Neuro+3 Study*

G.Force[1], I.Ghout[2], E.Bouaziz-Amar[3], K.Dorgham[4], D.Marigot-Outtandy[5], D.Troisvallet[6], V.Hahn[7], H.Defferrière[5], N.Darchy[1], J.Ropers[2], J.L.Laplanche[3], C. Delaugerre[8], G.Peytavin[9], G.Carcelain[10], P.deTruchis[5]

[1]IHFB Levallois; [2]APHP CHU A.Paré; [3]APHP CHU Lariboisière; [4]APHP CHU Pitié-Salpêtrière; [5]APHP CHU R.Poincaré; [6]CH Gonesse; [7]CH Ste-Anne; [8]APHP CHU St-Louis; [9]APHP CHU Bichat; [10]APHP CHU R.Debré; Paris, France

Contacts:
gilles.force@ihfb.org;
p.de-truchis@aphp.fr;

Background: HIV-associated neurocognitive disorders (HAND) are frequent despite plasma virological suppression on ART. Lower CNS Penetration Effectiveness (CPE) score is associated with higher risk of HIV replication in CSF[1] and HAND[2] and better CPE is known to prevent HAND[3]. In ARV controlled patients, ARV therapy optimization based on CSF HIV genotypes and CPE led to cognitive improvement and HIV control in CSF in retrospective studies[4,5]; in two randomized controlled trials, HAND improvement was associated with CPE intensification (13 patients[6] with high-CPE arm (p=0.057) and 14 patients[7] with maraviroc (MVC) intensification (p<0.05)), but small size of studies precludes drawing strong conclusions.

Study Design: We designed Neuro+3, an open-label prospective study of ARV intensification with higher CPE score in plasma HIV controlled patients, to test the hypothesis that HAND can be explained by residual HIV replication in CNS. Patients were screened with BREF and/or modified-HIV Dementia Scale to undergo a battery of 10 tests and 6 domains studied: Grooved Pegboard d&nd, Verbal Fluency, CVLT, Digit Span, PASAT, Digit Symbol, TMT A&B, Wisconsin Card Sorting Test. Raw test scores were converted to obtain a Global Deficit Score[8] (GDS) and patients were classified into HAND levels[9] (i.e. *Asymptomatic Neurocognitive Impairment* (ANI); *Mild Neurocognitive Disorder* (MND); *HIV-Associated Dementia* (HAD)) using the Cognitive Complaint Questionnaire (CCQ) and assessment scores. Brain MRI and CSF exam were performed to detect other neurological diseases. Plasma and CSF ultrasensitive HIV-RNA, inflammatory markers (neopterin (nM/L), sCD14, MCP-1, IP-10), neuronal damage marker NFL, and ARV drug concentrations were obtained at baseline and follow-up. After inclusion, current ARV was changed to a new combination with enhanced CPE ≥+3 (total CPE score ≥ 9) and same battery of tests was performed 48 and 96 weeks after ARV intensification. CPE score was corrected by cumulative genotype[10] and analyzed with ANRS algorithm.

Inclusion Criteria: informed consent; age between 18 and 65; plasma HIV-RNA <50 copies/mL during the past year; at least 2 altered neurocognitive domains with at least one standard deviation.

Exclusion Criteria: acute vascular or infectious disease of the CNS; positivity for HBsAg or HCV serology; drug or alcohol abuse; vitamin B deficiency; psychiatric disorders; only one altered ability domain; severe depression BDI II ≥29.

ARV Intensification: ARV strategies to improve CPE score were MVC addition (10 pts), addition or switch with integrase inhibitor (21 pts), DRV/r (6 pts), NNRTI (6 pts), and/or NRTI with abacavir (ABC) (4 pts). Intensification therapy consisted to change one drug class (1DC) in 18 patients, 2DC in 12 patients, and 3DC in one. After ARV intensification, 7 patients had new CPE score remaining <9 and ΔCPE score was only ≤+2 for 3 patients, 2 patients never took their new treatment regimen, thus only 22 patients had effective CPE ≥ 9 and 26 patients had ΔCPE score ≥+3.

Figure 1: Evolution of cognitive results with HAND Classification

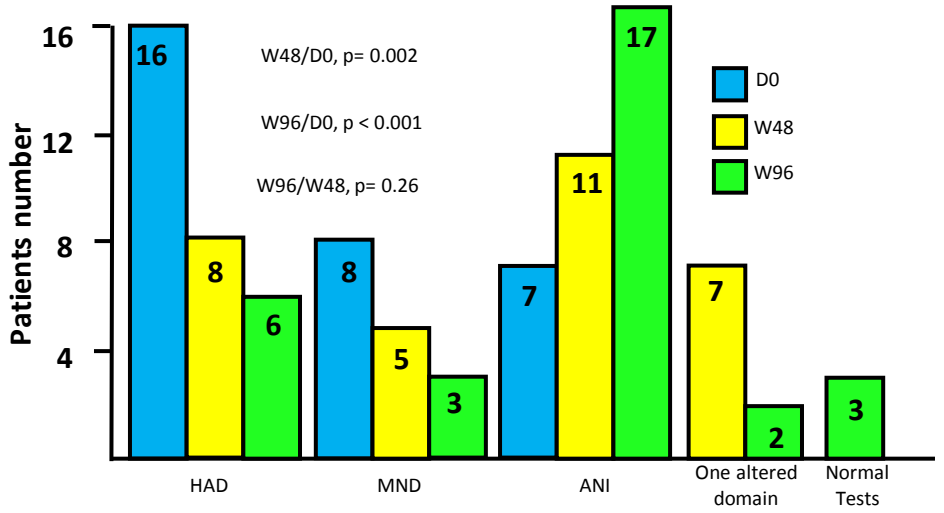

Patients number
16
12
8
4
W48/D0, p= 0.002
W96/D0, p < 0.001
W96/W48, p= 0.26
D0
W48
W96
16
8
6
8
5
3
7
11
17
7
2
3
HAD
MND
ANI
One altered domain
Normal Tests

CPE score of major drugs:
+4: DTG; NVP
+3: MVC; RAL; DRV/r; LPV/r; ETR; EFV; ABC; FTC
+2: EVG/c; ATV/r; RPV; 3TC **+1:** TDF

References:
1.Letendre, Arch.Neurol.2008
2.Smurzynski, AIDS 2011
3.Vassallo, AIDS 2014
4.Canestri, CID 2010
5.Peluso, AIDS 2012
6.Ellis, CID 2014
7.Gates, AIDS 2016
8.Carey, J.Cl.Exp.Neuropsy 2009
9.Antinori, Neurology 2007
10.Fabbiani, Antivir.Therap. 2015
11.Grant, Neurology 2014
12.Gott, PlosOne 2017
13.Winston, CID 2015
14.Zhang, Brain Res. 2012

Figure 2: Median cognitive change from D0 to W96:

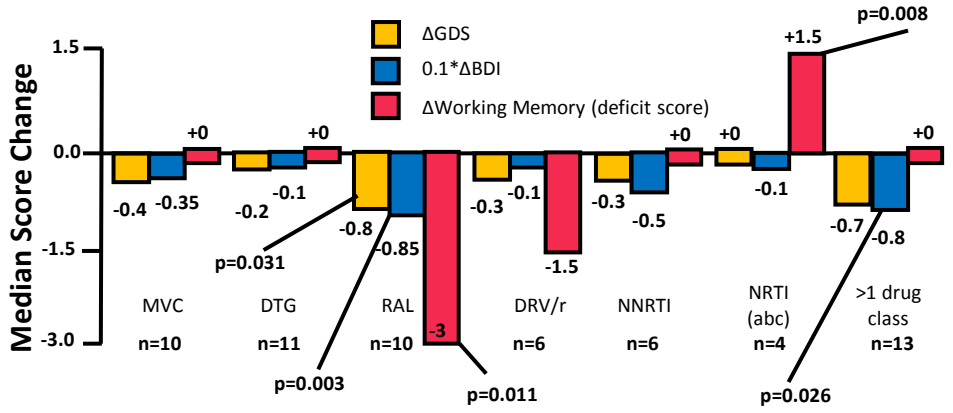

Median Score Change
1.5
0.0
-1.5
-3.0
ΔGDS
0.1*ΔBDI
ΔWorking Memory (deficit score)
+0
-0.4
-0.35
+0
-0.2
-0.1
-0.8
-0.85
-3
p=0.031
p=0.003
p=0.011
-0.3
-0.1
-1.5
-0.3
-0.5
+0
+0
-0.1
+1.5
p=0.008
-0.7
-0.8
+0
p=0.026
MVC n=10
DTG n=11
RAL n=10
DRV/r n=6
NNRTI n=6
NRTI (abc) n=4
>1 drug class n=13

Table 1: Baseline Characteristics

31 patients	Median results [IQR]
Age (years); Sex	55 [47-58]; 26 men, 5 women
Framingham Score	12 [7-20]
Educational level (years)	11 [9-14]
Duration (years) since: HIV Diagnosis	19 [8-24]
Last ART without change	3,3 [2,1-5,3]
Undetectable HIV-RNA	6,6 [5,2-9,1]
CD4 count (/mm3): Nadir/At baseline	165 [55-281] / 619 [396-773]
Plasma HIV-RNA: VL >5copies/mL	15 (48%)
Median Positive VL	13 copies/mL [7-18]
CSF HIV-RNA: VL >5copies/mL	9 (29%)
Median Positive VL	41 copies/mL [10-48]
CPE at baseline	6 [4-7]
Patients with CPE ≤6	18 (58%)
CPE after intensification	10 [9-11]
Δ CPE score	4 [3-4]

Figure 3: Cognitive Improvement according to ΔCPE score

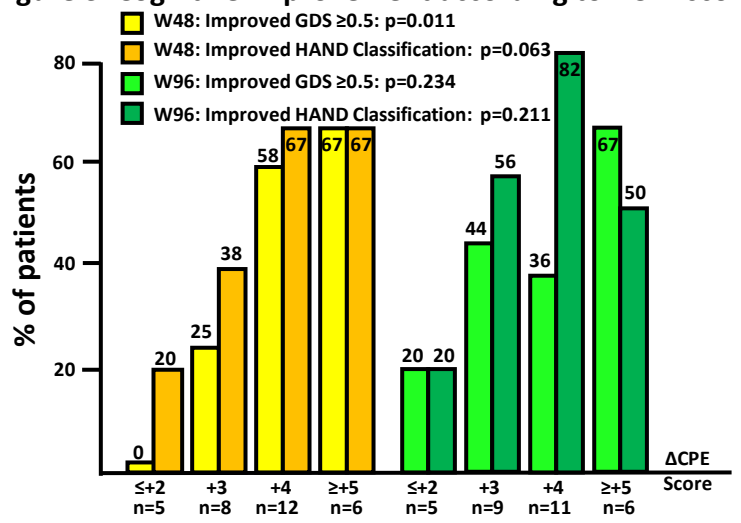

W48: Improved GDS ≥0.5: p=0.011
W48: Improved HAND Classification: p=0.063
W96: Improved GDS ≥0.5: p=0.234
W96: Improved HAND Classification: p=0.211
% of patients
80
60
40
20
0
20
25
38
58
67
67
67
20
20
44
56
36
82
67
50
≤+2 n=5
+3 n=8
+4 n=12
≥+5 n=6
≤+2 n=5
+3 n=9
+4 n=11
≥+5 n=6
ΔCPE Score

Results and Discussion: 63 HIV patients were screened and 31 enrolled, without any lost to follow-up. Neurocognitive improvement (ΔGDS ≥0.5 or/and HAND classification improved after 96 weeks) was observed in 22 patients (71%), an large difference compared to prospective cohorts[11,12] that showed worsening results over time. Baseline Characteristics are shown in Table 1. Evolution of cognitive results are presented in Figure 1 and Table 2, regarding each ARV or drug class in Figure 2. We observed best results in patients with more than one drug class change (>1DC) for ΔLearning Memory deficit score intensity (p=0.04) and ΔBDI II score. All patients maintained undetectable HIV VL in plasma, thirteen patients had positive CSF VL at any time, 9/31 at D0, 0/11 at W48 and 6/26 at W96, with no correlation between CSF VL and cognitive change. Evolutions of inflammatory markers are presented in Table 2, with evolution according to effective CPE ≥9 (Table 3) and to ΔGDS (Table 4).

Table 2: Evolution of cognitive and biologic markers for all 31 patients

Median [IQR]	Baseline	W48	W96	P (W48/B)	P (W96/B)
GDS	1,4 [0,8-2,2]	0,8 [0,6-1,6]	1,0 [0,6-2]	**0,012**	**0,009**
BDI II score	14 [6-25]	8 [3-15]	10 [3-17]	**0,008**	0,07
CCQ score	4 [2,5-5]	2 [0-4]	1 [0-3]	**<0,001**	**<0,001**
Altered domain	5 [3-5]	3 [2-5,5]	3 [2-5]	**0,012**	**0,005**
Neopterin (Pl ; CSF)	8 [6-13] ; 6 [5-8]	8 [7-12] ; 7 [5-8]	10 [7-12] ;5[5-7]	0,41 ; 0,20	0,34; 0,40
sCD14 Plasma	1600[1450-1920]	1840[1600-2025]	2050 [1808-2375]	**0,043**	**0,002**
CSF (ng/mL)	99 [76-121]	88 [60-96]	93 [69-103]	0,13	**0,032**
MCP-1 Plasma	198 [154-260]	240 [166-283]	230 [170-274]	0,11	0,67
CSF (pg/mL)	516 [416-590]	591 [456-689]	467 [383-628]	**0,033**	0,47
IP-10 Plasma	284 [233-571]	383 [204-587]	411 [199-584]	0,32	0,72
CSF (pg/mL)	654 [395-781]	579 [503-938]	525 [272-893]	0,77	0,48
NFL Plasma	12 [9-19] ; 700	14 [11-22]; 756	16 [12-23]; 913	**0,019**	**0,017**
CSF (pg/mL)	[536-950]	[705-853]	[634-1087]	0,9	0,1

Median plasma neopterin and IP-10 decrease in patients with GDS improvement and were stable in CSF. Median sCD14 significantly increased in plasma but decreased in CSF at W96. Despite a large correlation between NFL_{pl} and NFL_{csf} (rho=0.47, p=0.012), median NFL was significantly enhanced in plasma and stable in CSF at W96.

Factors associated with GDS improvement were at W48: ΔCPE score (p=0.035)(Figure 3); at W96, plasma neopterin decrease (p=0.006), plasma IP-10 decrease (p=0.006), RAL intensification (p=0.031), BDI II score decrease (p=0.008).

Discussion: Arguments for a link between cognitive improvement and Neuro+3 intervention were the positive relationship between cognitive improvement and ΔCPE score, better results for >1DC than 1DC change, significant difference in patients with effective CPE ≥9, and inflammatory markers decrease. Limitations of Neuro+3 study are small number of patients, non-randomized design, possible confounding factors such as depression and high cardiovascular risk but we observed same GDS improvement in different groups, suppression of potentially neurotoxic treatments such as EFV[13] or NRTI[14] (but only 7 patients concerned in our study: one switched EFV and GDS improved, 6 had a NRTI number decrease and improved GDS in 50%, result similar to the 5 patients with NRTI intensification (p=0.16)).

Table 3: Cognitive and biologic markers evolution according CPE ≥9 or not

Median [IQR]	Evolution at W96/D0 with CPE ≥9 (n=22)	Evolution at W96/D0 with CPE <9 (n=9)	p
Δ GDS	-0,4 [-0,8;-0,1]	0 [-0,2;+0,2]	**0,018**
Δ BDI II score	-5 [-11;-1]	1 [-1;+5]	**0,029**
Δ CCQ score	-1,5 [-4;0]	-2 [-2;-1]	1,0
Δ Altered domain	-1 [-3;0]	0 [-1;+1]	**0,025**
Δ Neopterin (Pl ; CSF)	0 [-2;+2]; -0,3 [-2;0,8]	1,3 [-1;5]; -0,4 [-1;0,3]	0,18; 0,86
Δ sCD14 (Pl ; CSF)	510[250;763]; -9[-35;1]	430[313;628];-4[-28;0]	0,64; 0,70
Δ MCP-1 (Pl ; CSF)	-18[-48;21]; -39[-50;38]	41 [-6;146]; 9[-75;84]	0,06; 0,78
Δ IP-10 (Pl ; CSF)	-28[-105;1];-78[-122;56]	57[7;211];-7[-131;280]	**0,028**; 0,31
Δ NFL (Pl ; CSF)	2,6 [1;6] ; 50 [-84;137]	2,9 [1-6]; 112 [54;311]	0,60; 0,27

Table 4: Biologic markers evolution according ΔGDS

Median [IQR]	Improved GDS at W96/D0 with ΔGDS >0,1 (n=19)	Failed GDS at W96/D0 with ΔGDS ≤0,1 (n=12)	p
ΔNeopterin (Pl ; CSF)	-0,4 [-1,9;1,4]; -0,5 [-2;0,5]	1,9 [0;4,7]; 0,1 [-0,4;0,8]	**0,028**; 0,21
Δ sCD14 (Pl ; CSF)	510 [233;708];-28 [-44;-4]	415 [285;650]; 0 [-4;1]	0,72; **0,05**
Δ MCP-1 (Pl ; CSF)	12 [-44;59]; -39 [-70;40]	-12 [-36;38]; 9 [-47;71]	0,79; 0,49
Δ IP-10 (Pl ; CSF)	-28 [-134;3];-82[-255;74]	77[-23;227];-7 [-61;99]	**0,03**; 0,31
Δ NFL (Pl ; CSF)	2,8 [1,5;7] ; 112 [11;294]	1,8 [-1;5,5]; 49 [-80;77]	0,57; 0,30

Conclusion: In our study in ARV controlled patients, CPE score intensification is correlated with better cognitive results, as in studies with 4 domains or more explored[6,7]. Despite its small sample size, our study give arguments for residual CNS HIV replication related to neurocognitive disorders in HAND. ARV intensification could lead to a better control of CNS HIV reservoir. Positive CSF VL could reflect compartmentalization intensity, but the majority of our patients had negative CSF VL. Thus, after eliminating confounding factors, we recommend ARV intensification strategy with high CPE score, and 2 drug class switch, to improve neurocognitive disorders in patients with HAND.

Acknowledgments: We are grateful to Lucette Cysique and Scott Letendre for their expert advices, and thank all patients, investigators, the monitoring team and staff who participated in Neuro+3 study.

Funding: This study was funded by Abbvie France.