Poster # 0744

Conference on Retroviruses and Opportunistic Infections CROI 2019
Mar 4-Mar 7, 2019
Seattle, Washington

Corresponding authors:
J. Sibiude (jeanne.sibiude@aphp.fr)
J. Warszawski (josiane.warszawski@inserm.fr)

NO INCREASE IN BIRTH DEFECTS IN INFANTS EXPOSED TO INTEGRASE INHIBITORS AT CONCEPTION

Jeanne Sibiude[1,2,3], Jérôme Le Chenadec[4], Laurent Mandelbrot[1,2,3], Stéphane Blanche[5,6], Catherine Dollfus[7], Nathalie Lelong[8], Elisa Arezes[1, 9], Lamya Ait Si Semi[1], Sophie Matheron[10], Christine Rouzioux[5,6], Josiane Warszawski[4,9,11], and Roland Tubiana[12,13], for the ANRS CO1-French Perinatal Cohort Study

[1]AP-HP Hôpital L. Mourier, Colombes; [2]UMR 1137, INSERM, IAME; [3]Univ Paris-Diderot, Paris; [4]CESP INSERM U1018, Le Kremlin-Bicêtre; [5]AP-HP Hôpital Necker-Enfants Malades, Paris; [6]Univ Paris-Descartes, Paris; [7]AP-HP Hôpital Trousseau, Paris; [8]Inserm, UMR S953, Université Paris 6, Paris; [9]Univ Paris Sud/Paris Saclay, Le Kremlin-Bicêtre; [10]AP-HP Hôpital Bichat, Paris; [11]AP-HP Hôpital Bicêtre, le Kremlin-Bicêtre; [12]Univ Paris-Sorbonne, Paris [13]AP-HP Hôpital Pitié Salpétrière, Paris, FRANCE

CESP - Inserm U1018

Institut national de la santé et de la recherche médicale

INSERM CESP 1018 - Hôpital de Bicêtre AP-HP
82 rue du Général Leclerc
94276 LE KREMLIN BICETRE Cedex
Tél. : 33 1 49 59 53 05/10 Fax : 33 1 49 59 53 00

OBJECTIVE

To compare birth defect rates and other maternal or neonatal outcomes between integrase-inhibitors (InSTI) exposed mother-infant pairs and matched non-InSTI-exposed pairs.

PATIENTS AND METHODS

The National French Perinatal Cohort (EPF, ANRS CO1/CO11)

Prospective national cohort including all pregnant women with HIV and their children in 90 French centres since 1986.
CO1 component of cohort records detailed information during the pregnancy, including immuno-virological data ; CO11 is simplified and represents 1/3 of the cohort.

Study population: Inclusion of all mother-infant pairs exposed to InSTI, categorized into 3 groups: (G1) ongoing at conception, (G2) initiated during pregnancy, as first-line regimen, and (G3) initiated during pregnancy, as second-line regimen.
Matching: Within each group, we matched 1:1 an InSTI-unexposed infant according to other drugs, ethnicity, center, year of delivery, and gestational age at ART-initiation. InsTI exposed women who did not receive PI or NNRTI were matched to women who received darunavir, with the same other drugs.
Statistical analysis: We compared birth defect rates between the 3 InSTI-exposed groups and, for each group, with the respective matched group, using chi2 and McNemar tests.

Table 1 – Baseline characteristics according to exposure group

Maternal baseline characteristics	Exposed at conception G1 N=301		InSTI as 1st line during pregnancy G2 N=183		InSTI as 2nd line during pregnancy G3 N=324		
	n	%	n	%	n	%	p[a]
Age (years)							
<25	21	**7.0**	33	**18.0**	32	**9.9**	<0.01
25-34	156	**52.0**	114	**62.3**	195	**60.2**	
≥35	123	**41.0**	36	**19.7**	97	**29.9**	
Nulliparous	79	**26.3**	45	**25.1**	66	**20.6**	0.21
Geographical origin							
Metropolitan France	50	**16.6**	11	**6.0**	18	**5.3**	<0.01
Sub-saharan Africa	198	**65.8**	133	**72.7**	242	**75.6**	
Other	53	**17.6**	39	**21.3**	60	**18.8**	
HIV diagnosis this pregnancy	0	**0.0**	75	**41.9**	251	**77.9**	<0.01
> 10 Years HIV-infected*	76	**40.4**	8	**8.6**	52	**23.1**	<0.01
Viral load at delivery > 50 cp/mL	28	**11.0**	61	**35.9**	105	**34.7**	<0.01
CD4 at delivery < 200/mL	9	**4.3**	24	**16.3**	38	**14.0**	<0.01

* Only available for CO1 component N=506, [a]: Chi2 test

Fig 1 – Selection of study population

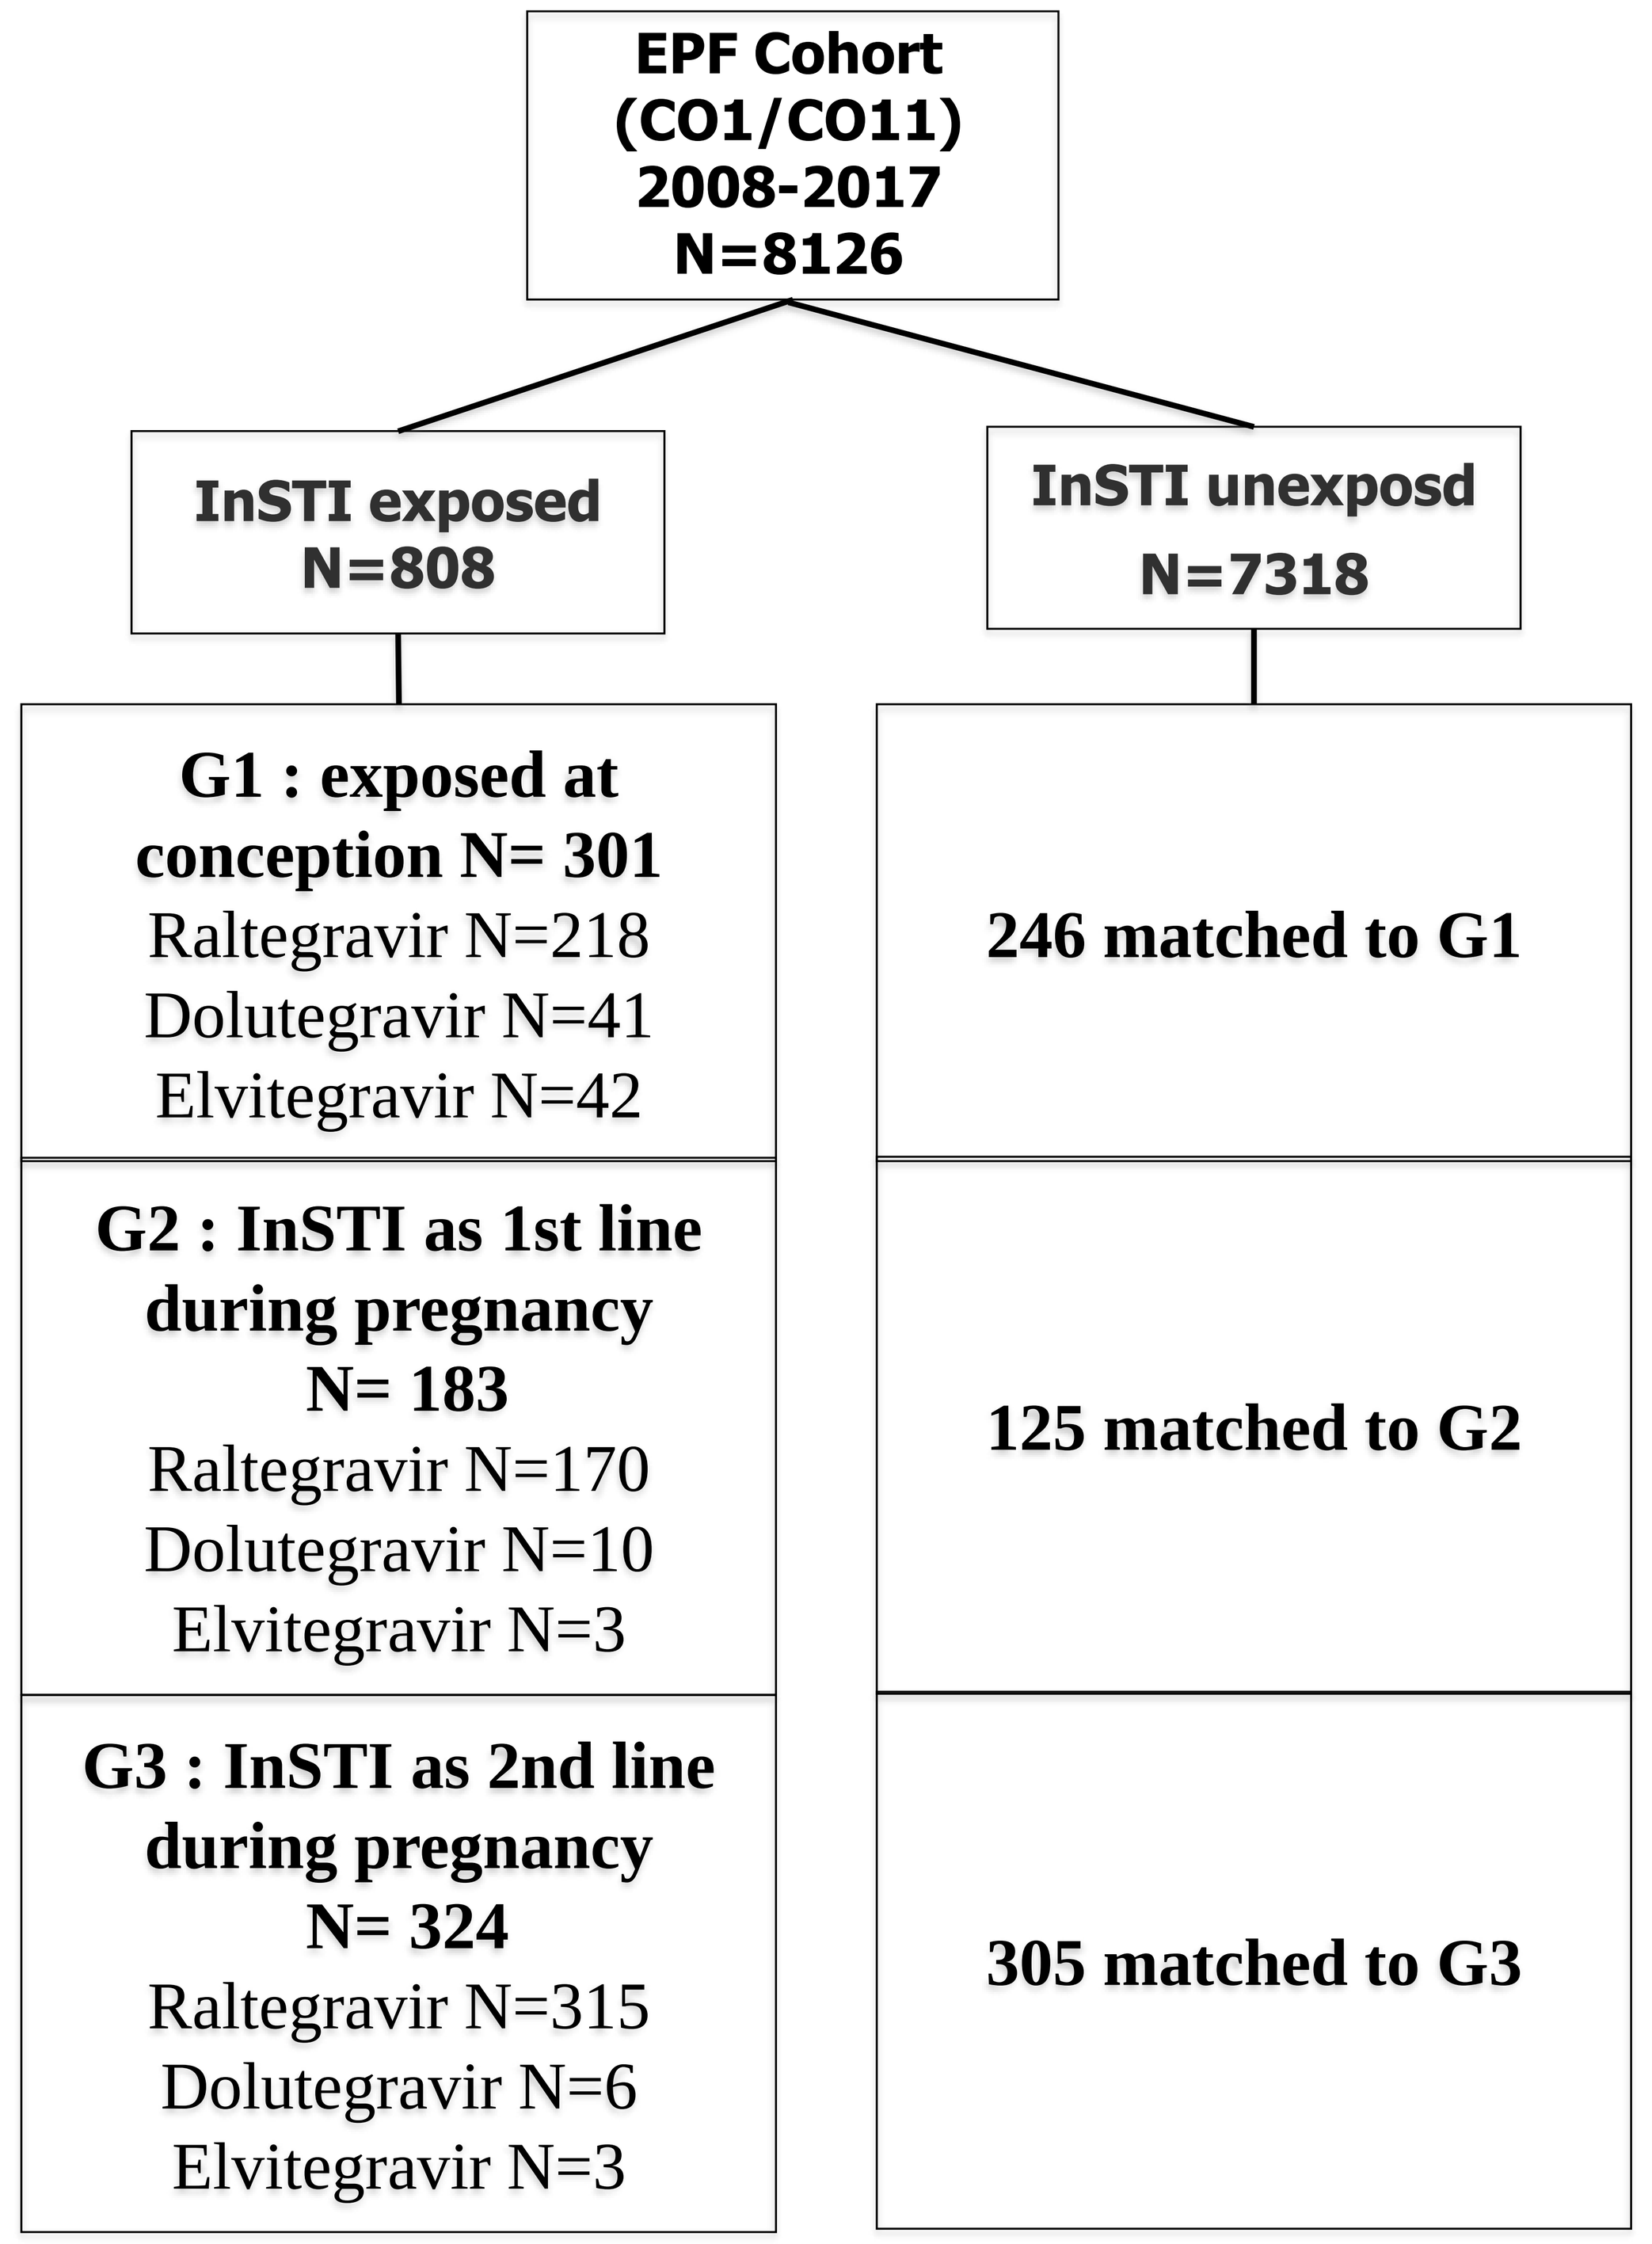

EPF Cohort (CO1/CO11) 2008-2017 N=8126
InSTI exposed N=808
InSTI unexposd N=7318
G1 : exposed at conception N= 301
Raltegravir N=218
Dolutegravir N=41
Elvitegravir N=42
246 matched to G1
G2 : InSTI as 1st line during pregnancy N= 183
Raltegravir N=170
Dolutegravir N=10
Elvitegravir N=3
125 matched to G2
G3 : InSTI as 2nd line during pregnancy N= 324
Raltegravir N=315
Dolutegravir N=6
Elvitegravir N=3
305 matched to G3

Table 2 – Perinatal outcomes according to exposure group

Perinatal outcomes	Exposed at conception G1 N=301		InSTI as 1st line during pregnancy G2 N=183		InSTI as 2nd line during pregnancy N=324		
	n	%	n	%	n	%	p[a]
Birth defect	18	**5.8**	5	**2.7**	9	**2.7**	0.09
Stillbirths	7	**2.4**	2	**1.1**	1	**0.3**	0.07
Preterm Birth	50	**16.8**	22	**12.1**	47	**14.6**	0.36
Birthweight < 3rd centile	9	**3.0**	6	**3.4**	20	**6.2**	0.13
Length < 3rd centile	12	**4.4**	9	**5.5**	14	**4.5**	0.86
Head circumference <3rd centile	8	**2.9**	8	**4.8**	11	**3.5**	0.56

[a]: Chi2 test

Table 3– Perinatal outcomes: comparison of InSTI-exposed and matched pregnancies

Perinatal outcomes	InSTI-exposed matched		InSTI-unexposed matched			InSTI-exposed unmatched	
	n	%	n	%	p[a]	n	%
Exposed at conception (G1)							
	N=246		N=246			N=55	
Birth defect	14	**5.7**	7	**2.9**	0.13	4	**7.3**
Stillbirth	6	**2.5**	6	**2.5**	1	1	**1.9**
Preterm birth	41	**16.8**	39	**16.1**	1	9	**17.0**
Unexposed to any ART at conception. InSTI as 1st line during pregnancy (G2)							
	N=125		N=125			N=58	
Birth defect	4	**3.2**	10	**8.0**	0.12	1	**1.7**
Stilllbirth	2	**1.6**	1	**0.8**	0.57	0	**0.0**
Preterm birth	16	**12.8**	14	**11.2**	0.70	6	**10.5**
InSTI as 2nd line during pregnancy (G3)							
	N=305		N=305			N=19	
Birth defect	8	**2.6**	14	**4.6**	0.21	1	**5.6**
Stilllbirth	0	**0.0**	0	**0.0**	1	1	**5.3**
Preterm birth	45	**14.8**	41	**13.5**	0.65	2	**10.5**

[a]: McNemar test for matched data

Table 4 – Description of birth defects according to exposure

InSTI-Exposure at conception	N	InSTI-Exposure at conception	N
Raltegravir		Dolutegravir	
Chromosomal abnormalities (T21 N=1, T18, N=2)	3	Chromosomal abnormalities	1
Polydactyly	4	Patent ductus arteriosus	1
Patent ductus arteriosus	2	Elvitegravir	
Anomalous pulmonary venous drainage	1	Polydactyly	1
Coartctation of the aorta	1		
Corpus callosum agenesis	1		
VACTERL syndrome	1		
Hip dislocation	1		
Posterior urethral valves	1		

RESULTS

Since 2008, 808 women were exposed to InSTI, during pregnancy, 301 were exposed at conception (G1), 183 started an InSTI-based treatment as 1st line during pregnancy (G2) and 324 were treated with an InSTI-based treatment as second-line (intensification) (G3).

Women from the 3 groups differed significantly : women on InSTI at conception were older, and had been living with HIV for a longer time (Table1).

The rates of birth defects and of stillbirths tended to be higher among women on InSTI at conception vs InSTI later in pregnancy, without reaching significance. Other perinatal ouctomes did not differ (Table 2).

Exposed and matched InSTI-unexposed groups did not differ in rates of birth defects, stillbirths or preterm birth (Table3).

Among birth defects reported in women InSTI-exposed at conception, we found no neural tube defects (Table 4).

CONCLUSION

We found no evidence of a higher birth defect rate among 301 women exposed to InSTI at conception, mostly exposed to raltegravir, however in the current context, surveillance must be pursued for this class of ART.

ABSTRACT

Background : Integrase inhibitors (InSTI) are increasingly used by HIV-infected women during pregnancy. Following an alert on the association of dolutegravir with neural tube defects, we evaluated the risk of birth defects in case of exposure to this antiretroviral class.
Methods The French Perinatal Cohort is a multicenter national cohort including all HIV-infected women in 90 maternities. We studied all mother-infant pairs exposed to InSTI, categorized into 3 groups: (G1) ongoing at conception, (G2) initiated during pregnancy, as first-line regimen, and (G3) initiated during pregnancy, as second-line regimen. Within each group, we matched 1:1 an InSTI-unexposed infant according to other drugs, ethnicity, center, year of delivery, and gestational age at ART-initiation. InsTI exposed women who did not receive PI or NNRTI were matched to women who received darunavir, with the same other drugs. We compared birth defect rates between the 3 InSTI-exposed groups and, for each group, with the respective matched group, using chi2 and McNemar tests.
Results : Overall, 301 women were exposed to InSTI at conception (G1): 218 exposed to raltegravir, 41 to dolutegravir and 42 to elvitegravir. Rates of birth defects for InSTI-exposed infants at conception (G1: 5.8% 18/301) did not significantly differ from those of InSTI-exposed infants of the two other groups: 2.7% (5/183) in G2 and 2.7% (9/324) in G3, p=0.09. There was no neural tube defect among infants exposed to InSTI at conception. There were only two birth defects among the 41 infants exposed to dolutegravir (a case of Down syndrome, and a persistant ductus arteriosus).When restricting to matched infants, birth defect rates in G1 were not significantly different from the matched InSTI-unexposed group (5,7%, 14/246 vs 2,9%, 7/246, respectively, p=0.13). The EUROCAT types of birth defects were similar for InSTI-exposed at conception and matched infants. There was no difference in stillbirth rates (2,5% vs 2,5%, p=1), nor in preterm birth rates (16,8% vs 16,1%, p=1) between pregnancies exposed at conception and the matched pregnancies. Among women exposed at conception, 65% were still receiving InSTI at delivery. Similarly, there was no difference in birth defect rates between InSTI-exposed infants in G2 and G3 and the matched unexposed infants.
Conclusion We found no evidence of a higher birth defect rate among 301 women exposed to InSTI at conception, mostly exposed to raltegravir, however in the current context, surveillance must be pursued for this class of ART.

Many thanks to all the centers participating in the French Perinatal Cohort: APHP Hôpital Louis Mourier, Colombes, France (Laurent Mandelbrot, Catherine Crenn-Hebert*, Corinne Floch-Tudal*, Fabienne Mazy, Marine Joras, Françoise Meier, Emmanuel Mortier, Catherine Briquet, , Houria Ichou, Laurence Marty, Hélène Jabbarian); APHP Hôpital Beaujon, Clichy, France (Pierre-François Ceccaldi*, Maïa Banige, Agnès Villemant Uludag, Virginie Zarouk, Agnès Lefort, Mariam Ben Salah); Hôpital Sainte Musse, Toulon, France (Gilles Hittinger*, Jean-Marc Chamouilli, Christian Burle, Alain Lafeuillade, Gisèle Philip,Véronique Lambry); CHG Marechal Joffre, Perpignan, France (Marie Medus*, Germaine Bachelard, Martine Malet); CHU Caremeau, Nîmes, France (Joëlle Dendale-Nguyen*); CHD les Oudairies, La Roche sur Yon, France (Thomas Guimard*, Karine Guimard, Jean-Pierre Brossier, Philippe Perré, Jean-Luc Esnault, Olivier Aubry, Sophie Leautez-Nainville, Valerie Bonnenfant, , Laeticia Laine); Centre Hospitalier William Morey, Châlon sur Saone, France (Sandrine-Anne Martha*, Elise Maurel, Michel Françoise, Muriel Barat, Patricia Murger); Centre Hospitalier, Vernon, France (Mahfoud Rouha*, Marc Lévy, Philippe Lumbroso, Alain Checoury, Osseni Sahadatu, Johnson)); Centre Hospitalier Intercommunal de Cornouaille, Quimper, France (Pascale Perfezou*, Gilles Blondin, Jean-Charles Duthé); Centre Hospitalier Universitaire, Brest, France (Séverine Ansart*, Luc De Saint Martin*, Philippe Le Moine, Jean-Charles Duthé); Centre Hospitalier, St Brieuc, France (Corinne Daniel*, Christian Calvez, Emmanuelle Boutaric, Jennifer Rohan); Centre Hospitalier Universitaire, Rennes, France (Cédric Arvieux*, Estelle Bauville, Christelle Dupre, Pascal Lotton, Enora Ouamara-digue); Centre Hospitalier Bretagne Atlantique, Vannes, France (Yves Poinsignon*, Marie Goussef, Anne Grelier, Gaetane Mousset, Corinne Cudeville, Virginie Mouton-Rioux); Centre Hospitalier de Bretagne Sud, Lorient, France (Mathilde Niault*, Isabelle Belzic, Philippe Moreau, Marie-Françoise Le Coz, Odile Luycx Vaillant, Anne Guerin-Duplessy, Virginie Mouton-Rioux, Philippe De Morel); Centre Hospitalier de la région d'Annecy, Annecy, France (Virginie Vitrat*, Didier Tardif, Jacques Gaillat, Anne Vanderbergh, Suzanne Braig, Gaelle Clavere); Centre Hospitalier Intercommunal, Montfermeil, France (Marion Dehlinger-Paul*, Khaled Mohamed, Marie Echard, Michel Camus, Catherine Mulard, Marie-Agnès Fontelonga); Centre Hospitalier Intercommunal, Montreuil, France (Brigitte Heller-Roussin*, Cécile Winter, Marion Challier, Elise Debruyne); APHP Hôpital Cochin-Port Royal, Paris, France (Valerie Marcou*, Ghislaine Firtion, Emmanuelle Pannier*, Myriam Costa, Odile Launay, Dominique Salmon Ceron, Touria Belkacem, Youcef Bajawi, Valérie Raynal Aubret, Danièle Rivaux); APHP Hôpital Bichat, Paris, France (Sophie Matheron*, Neila Elaoun, Lahcene Allal, Sandrine Djoubou, Djamila Rahli, Agnès Bourgeois Moine, Morgane Valentin, Florence Damond, Virginie Huri, Valérie Vivier, Fatma Ait Yahia); Centre Hospitalier Intercommunal, Créteil, France (Valérie Garrait*, Isabelle Hau*, Claudine Touboul, Lanto Ratsimbazafy, Emilie Boiron, Brigitte Elharrar, Laurent Richier); Hôpital de la Croix Rousse, Lyon, France (Laurent Cotte*, Jean-Marc Labaune, René-Charles Rudigoz, Corinne Brochier, Valérie Galvan, Stanislas Ogoudjobi); Centre Hospitalier Pellegrin, pauchart, Bordeaux, France (Christophe Elleau*, Camille Runel-Belliard*, Thierry Pistone, Hervé Fleury, Jacques Horovitz, Boris Sandler, Denis Roux, Jean-Marie Ragnaud, Pierre Chabanier, Jean-Luc Brun, Sandrine Delveaux); CHU Les Abymes, Pointe à Pître, France (Blandine Muanza*, Mama Doufari Diallo, Isabelle Lamaury, Marie-Thérèse Sow, Ketty Samar); Centre Hospitalier Général, Creil, France (Bénédicte Carpentier*, Zafer Osman, Etienne Dienga); Centre Hospitalier Général, Longjumeau, France (Hervé Seaume*, Sarah Ducrocq, Philippe Bailly-Salin,Christelle Dusart Da Silva, Isabelle Fayolas); Hôpital Paule de Viguier, Toulouse, France (Julie Abbal*, Caroline Simon-Toulza*, Véronique Truillet, Noëlle Bogner, Julie Chiabrando, Evelyne Armand); Centre Hospitalier de la Côte Basque, Bayonne, France (Claudine Cayla*); Centre hospitalier intercommunal, Villeneuve St Georges, France (Anne Chacé*, Isabelle Matheron, Laurent Richier, Joe Miantezila, Sandrine Bry); Centre Hospitalier Intercommunal, Poissy Saint Germain en Laye, France (Sophie Couderc*, Catherine Narcy); Centre Hospitalier Général, Fontainebleau, France (Corinne Routier*, Rania Nassar); Centre Hospitalier Robert Ballanger, Aulnay, France (Marie-Anne Bouldouyre*, Ahmed Zakaria, Hélène Dauphin, Céline Goissen, Marie Belloy, Jean-Luc Delassus, Véronique Favret, Céline Nemeth); Hôpital Hautepierre, Strasbourg, France (MariaLuisa Partisani*, Natacha Entz-werle, Bruno Langer, Françoise Uettwiller, Myriam Durand) ; Hôpital Civil, Strasbourg, France (MariaLuisa Partizani*, Christine Cheneau, David Rey, Edith Ebel, , Patricia Fischer) ; Centre médico-chirurgical Obstétrique, Strasbourg, France (Eric David, Christophe Vayssiere, Michèle Weil, Marie Paule Schmitt, Israël Nisand) ; Centre Hospitalier Victor Dupouy, Argenteuil, France (Philippe Genet*, Dominique Brault, Christine Allisy, Juliette Gerbe, Virginie Masse, Bouchra Wifaq, Laurence Courdavault, Petra Gabor, Nathalie Tordjeman); APHP Hôpital Tenon, Paris, France (Marie-Gisèle Lebrette*, Lise Selleret, Déborah Samama, Geneviève Vaudre); Centre Hospitalier Général, Saint-Denis, France (Pascal Bolot*, Marie-Aude Khuong-Josses, , Mahdi Amel, Stéphane Bounan, Christelle Nourry, Sabine Andris); APHP Hôpital Necker, Paris, France (Stéphane Blanche*, Marine Driessen*, Pierre Frange, Florence Veber, Alain Fisher, Christine Rouzioux, Véronique Avettand-Fenoel, Nizar Malaoui, Marie-Christine Mourey); Centre Hospitalier Sud Francilien, Evry Corbeil, France (Michèle Granier*, Alain Devidas*, Anne-Claire Donnadieu, , Adrien May, Amélie Chabrol, Pierre Chevojon, Chahrazede Bellahcene, Audrey Sanchez, Claire Malbrunot, Joelle Neizelien, Nouara Agher); Centre Médico-Chirurgical et Obstétrical, Schiltigheim, France; CHR American Memorial Hospital, Reims, France (Claire Pluchart*, Christine Rouger); APHP Groupe Hospitalier Pitié Salpêtrière, Paris, France (Roland Tubiana*, Marc Dommergues, Manuela Bonmarchand, Luminata Shneider, Fabienne Caby, Ruxandra-Oana Calin, Christine Blanc, Catherine Lupin, michèle Pauchard); Centre Hospitalier René Dubos, Pontoise, France (Mohamed Amine Yangui*, Didier Roca, Darina Todorova, Juliette Laurent, A Ferry, Martine Deschaud, Laurent Blum); APHP Hôpital Béclère, Clamart, France (Véronique Chambrin*, Philippe Labrune*, Laure Clech, Mariem Raho-Moussa); Centre Hospitalier Marc Jacquet, Melun, France (Isolde Pauly-Ravelly*, Thierry Jault, Soufiane Bouabdallah, Lydie Sanchez, Anita Sanchez); Centre Hospitalier Général, Evreux, France (Ama Johnson*, Agnès Louchard, Claude Allouche, Jean Paul Pathe); APHP Hôpital Jean Verdier, Bondy, France (Eric Lachassine*, Laurence Benoist, Vincent Jeantils, Catherine Delannoy, , Amélie Benbara, Lionel Carbillon, Anne Borgne, Laurence Moreau, Fabienne Picard); Centre Hospitalier de Meaux, Meaux, France (Leïla Karaoui*, Véronique Lefevre Elbert, Valérie Balaz); CHU de l'Archet, Nice, France (André Bongain*, Fabrice Monpoux*, Anne Deville, Eliane Galiba); Centre Hospitalier François Quesnay, Mantes La Jolie, France (Ahmed Jabbar*, Martine Joutel); CHU Hôpital Nord, Amiens, France (Jean-Luc Schmidt*, Nathalie Decaux); Hôpital de la Conception, Marseille, France (Ludovic Cravello*, Katia Errichiello); CHU de Brabois-Hôpital des Adultes, Vandoeuvre les Nancy, France (Claire Hubert*); APHP Hôpital Trousseau, Paris, France (Catherine Dollfus*, François Hervé, Marie-Dominique Tabone, Mary-France Courcoux, Guy Leverger, Gilles Kayem, , Aurélie Schnurgier, Aurore Jensen, Geneviève Vaudre); Hôpital Charles Nicolle, Rouen, France (Didier Pinquier*, Alexis Gromez , Gaelle Pinto-Cardoso,); APHP Hôpital Robert Debré, Paris, France (Albert Faye, Constance Borie*, Martine Levine*, Sophie Matheron, Erianna Bellaton Marouts, Christine Boissinot, Marion Caseris, Virginie Pommelet, Geneviève Morau, Sandrine Leveille, Marie Astride Boumediene, Dominique Garion); APHP Hôpital de Bicêtre, Le Kremlin-Bicêtre, France (Delphine Peretti*, Corinne Fourcade*, Marie Houllier, Ikram Jrad, Katia Bourdic, Sylvie Monnier); CHU Hôpital Jean Minjoz, Besançon, France (Catherine Chirouze*, Aurélie Proust, Odile Catteau, Quentin Gardiennet); CHU de Nantes, Nantes, France (Véronique Reliquet*, Cécile Brunet-Cartier*, Norbert Winer, Edouard Vaucel, Audrey Rodallec, Elisabeth Garnier-André); CHRU Hôpital du Bocage, Dijon, France (Claire Briandet*); CHRU Hôpital Clemenceau, Caen, France (Jacques Brouard*, Pascale Goubin, Gaël Beucher, Julia Dina); Centre Hospitalier de Lagny, Lagny, France (Arnaud Chalvon Demersay*, Sylvie Tassi, Gaelle Lavarenne); Hôpital André Mignot, Le Chesnay, France (Mandovi Rajguru*, Fabienne Messaoudi, Nathalie Carre, Mandavi Rajguru, Claire Bobrie-Moyrand, Pierre Foucaud); CHRU de Tours, France (Louis, Bernard*, Zoha Maakroun, Olivier Bourgault); Institut d'Hémato-Oncologie Pédiatrique, Lyon, France (Kamila Kebaïli*, Yves Bertrand, Véronique Alixe, Emeline Boyer); Hôpital Nord, Saint Etienne, France (Kareen Billiemaz*, Cécile Fanget, Véronique Ronat, Catherine Lesauder, Marie Laure Lavastre); Centre Hospitalier Général, Bastia, France (Alice Moulin*, Marie-France Turquini, Dominique Colombani, Danièle Belgodere); Centre Hospitalier Universitaire, Angers, France (Pascale Fialaire*, Stéphanie Proust, Sami Rehaiem); Centre Hospitalier Régional, Orléans, France (Louis Mesnard, Evelyne Werner*, Nathalie Dukiel, Baya Desmergers, Isabelle Blanc-Ruffat); APHP Hôpital Lariboisière, Paris, France (Barbara Maraux*, Anne Coursol, Julie Castaneda,Lise Etienne); CHR Arnaud de Villeneuve, Montpellier, France (Emmanuelle Vintejoux*, Muriel Lalande, Jacques Reynes, Michel Segondy, Nelly Guigue); Centre Hospitalier Général, Orsay, France (Christiane De Gennes*); Centre Hospitalier de Saint Martin, St Martin, France (Cyril Clavel*, François Cazassus, Véronique Walter); CHR Jeanne de Flandres, Lille , France (Françoise Mazingue*, Yamina Hammou, Marion Lagree, Odile Paquiez, Sophie D'angelo, Faiza Ajana, Laurence Boquet); Centre Hospitalier Dron, Tourcoing, France (Faïza Ajana); CHU - Maison de la Femme et de l'Enfant, Fort de France, France (Yves Hatchuel*, Imad Nahri, Jenny Zebelus); CHU Dupuytren, Limoges, France (Claire Genet*, Sophie Ducroix-Roubertou, Yves Aubrard, Anne Constanty, Pierre Weinbreck); Hôpital Intercommunal Sud Léman Valserine, Saint Julien, France (Emilie Piet*, Françoise Jacquier); Centre Hospitalier Saint Nazaire Cité Sanitaire, Saint Nazaire, France (Christophe Michaud*, Hassan Safwan, Arnaud Boutet, Carole Grand-Courault); Groupe Hospitalier Saint Joseph, Paris, France (Fanny Autret*, Fakher Habibi, Elie Azria); Centre Hospitalier Léon Binet, Provins, France (Mohamed Abdelhadi*); Centre Hositalier Andrée Rosemon, Cayenne, France (Narcisse Elenga*); Hôpital Calmette, Lille, France (Laurence Bocket, Françoise Taillet); Centre Hospitalier de Béziers, France (Gilles Palenzuela*, Redouane Khadly ,Danielle Pierronnet, Emmanuelle Dos-Santos); Centre Hospitalier de Narbonne (Selva David*); Hôpital Edouard Herriot, Lyon, France (Djamila Makhloufi*, Florence Brunel-Dalmas, Elisabeth Carbonnel-Delalande, Pierre Chiarello, Matthieu Godinot, Sylvie Gilbert); Hôpital Femme Mère Enfant, Bron, France (Jérôme Massardier*, Hélène Gauthier-Moulinier) ; Centre Hospitalier de la Basse-Terre, Guadeloupe (Elisabeth Fernandes*, Sata Ranaivojaona, Coralie Chevry)